Manchester Metropolitan University

e-space

Manchester Metropolitan University's Research Repository

Please cite the Published Version

Betts, A, Odeyemi, I, Devlin, N, Yeowell, G and Fatoye, F (2017) Addressing uncertainty in wound management using a modified Delphi methodology. [Conference or Workshop Item]

DOI: https://doi.org/10.1016/j.jval.2017.08.2342

Publisher: Elsevier

Downloaded from: https://e-space.mmu.ac.uk/619391/

Additional Information: This is an author accepted abstract of a paper presented at ISPOR's 20th Annual European Congress, November 4-8, 2017, Glasgow. Publication of full paper forthcoming.

Enquiries:
If you have questions about this document, contact openresearch@mmu.ac.uk. Please include the URL of the record in e-space. If you believe that your, or a third party's rights have been compromised through this document please see our Take Down policy (available from https://www.mmu.ac.uk/library/using-the-library/policies-and-guidelines)

Addressing uncertainty in wound management using a modified Delphi methodology

Betts, A. Odeyemi, I. Yeowell, G. Fatoye, F. Devlin, N.
Email: a.betts@mmu.ac.uk

Objective

To gain consensus surrounding uncertainty in using dressings to improve wound outcomes.

Background

Chronic wounds such as Diabetic Foot Ulcers (DFU) and Leg Ulcers (LU) are increasingly prevalent and are a financial burden on the healthcare system, a 2016 estimate of long lasting ulcers below the knee was 15 out of every 10,0000 people[1]. A 2010-11 estimate calculated that approximately £1 in every £140 of NHS spending is on foot ulcers or amputations each year[2].

Wound dressings are a mainstay of treatment, however, the availability of a wide variety of dressings coupled with a lack of specific guidance presents uncertainty. NICE NG19 states that clinicians are to use “dressings with the lowest acquisition cost appropriate to the clinical circumstances”[3]. Cochrane reviews highlight the lack of robust studies with high levels of evidence surrounding several dressing types [4, 5].

To address the uncertainty regarding the use of dressings on chronic wounds, a modified Delphi methodology expert panel, involving two iterations of email questionnaires, and one face to face meeting, was conducted to elicit expertise from a multidisciplinary group of experts.

Methods

- The modified process used for this study is shown below:

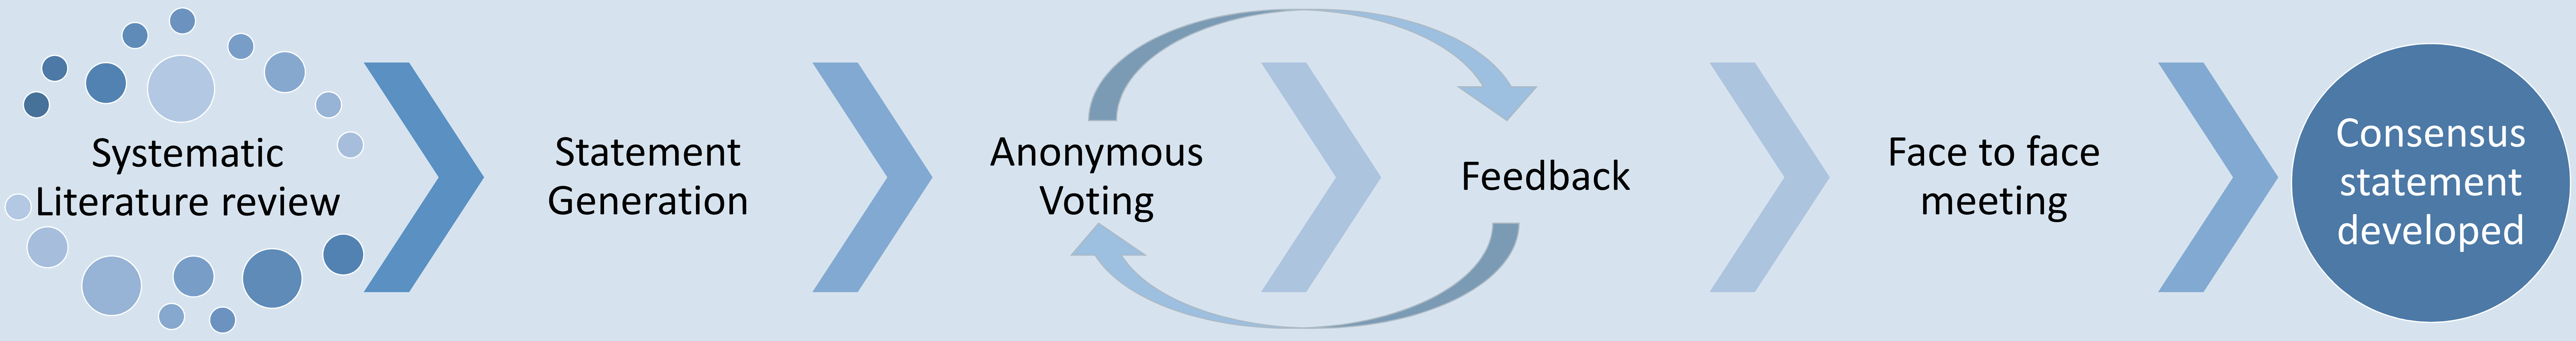
Systematic Literature review
Statement Generation
Anonymous Voting
Feedback
Face to face meeting
Consensus statement developed

- The consensus threshold was 80%, and participants could vote yes or no against the statements, confirming or rejecting them. Unconfirmed statements were modified according to the participants’ comments and resubmitted in the next round.

Results

The ten clinical experts on the panel represented Nursing, Tissue Viability, Podiatry, Surgery and Diabetology. Six technical experts representing Qualitative Research, Health Policy and Health Economics, were present to advise on the process, but did not have voting rights on the statements.

Due to the large number of statements confirmed before the final round, as shown in the table to the right, it was considered prudent to revisit comments on statements which had been confirmed with a level of 80-99%, in order to increase the level of consensus and ensure semantic clarity.

The final confirmed statements were used to create a larger consensus statement that had the agreement of the entire panel. This consensus statement is currently awaiting publication.

Round I	
Statements confirmed	38 (81%)
Statements unconfirmed	9 (19%)
Statements rejected	0 (0%)
Round II	
Statements confirmed	5 (56%)
Statements unconfirmed	4 (44%)
Statements rejected	0 (0%)
Face to Face meeting	
Statements confirmed	4 (100%)
Statements unconfirmed	0 (0%)
Statements rejected	0 (0%)

Discussion

The modified Delphi Methodology vs a traditional Expert Panel

Delphi Methodology	Expert Panel
The methodology is structured to place equal weight on the opinion of all panel members.	Unstructured expert panels or advisory boards can be led by dominant or more senior individual.
Iterative; multiple rounds of voting encourages individuals to reflect on their own opinions and knowledge in the context of feedback from others.	Usually a single meeting, individuals are encouraged to put forward their own opinions and not necessarily reach a consensus.
Participants are anonymous when they feed back their opinions.	Participants are not anonymous to one another.
Transparent methodology, the workbook is the basis for all discussions.	Unstructured method without controls on biases. Can allow for more freedom of discussion.

Strengths

- This study aimed to address uncertainties in clinical practice by developing a set of evidence-based statements, validated by experts.
- The systematic literature review reported using PRISMA guidelines and the use of a structured workbook to collect expert opinions allows for repeatability and validation of the results.
- Given the culture of regulatory scrutiny, using a Delphi methodology facilitated by an independent academic institution, protects the legitimacy of scientific exchange between Clinical experts themselves, and between the experts and the sponsoring manufacturers.

Limitations

- The binary voting system did not allow any ranking of the statements.
- As a result of working with opinions, a Delphi panel is subject to low levels of evidence classifications.

References:
1. Cullum N, Buckley H, Dumville J, et al. (2016) Wounds research for patient benefit: a 5-year programme of research. Programme Grants Applied Research. 4(13)
2. Kerr M, Rayman, G. Jeffcoate, W.J. (2014) Cost of diabetic foot disease to the National Health Service in England. Diabetic Medicine. 31(12)
3. NICE guideline (2015) Diabetic foot problems: prevention and management. NG19 [Online] [Accessed 24th October 2017] nice.org.uk/guidance/ng19
4. Dumville, J. C., Deshpande, S., O'Meara, S. and Speak, K.(2013) Foam dressings for healing diabetic foot ulcers. Cochrane Database of Systematic Reviews. (Issue 6. Article: CD009111)
5. Westby, M. J., Norman, G., Dumville, J. C., Stubbs, N. and Cullum, N. (2016). Protease-modulating matrix treatments for healing venous leg ulcers. Cochrane Database of Systematic Reviews. (Issue 12. Article: CD011918)

Source of funding:
This project was commissioned and funded by Urgo Medical UK and designed and executed by Manchester Metropolitan University.